Small nuclear RNA-Mediated Exon 51 Skipping AAV9 Gene Therapy for the Treatment of Duchenne Muscular Dystrophy

Rea Lardelli Markmiller, Greg Nachtrab, Claire Geddes, Takahiro Tadokoro, Daniel Knowland, Anindita Sarkar, Emily Miyoshi, Sheik Pasha, Daniela Roth, Alberto Carreño, Derek Reid, Oscar Diago, Shruti Vakharia, Bryan Almaguer, Lynn Nguyen, Yesol Go, Ronald Torres, Nandini Narayan, Hank Bradford, Jeannie Chew, Deepti Vashist, Gustavo Villegas, Rachel Adams, Haydee Gutierrez, Aaron Berlin, Amanda Tran, Ranjan Batra, John P. Leonard

Locanabio, Inc. 3545 John Hopkins Ct, San Diego, CA 92121

Duchenne Muscular Dystrophy

- Duchenne Muscular Dystrophy (DMD) is an X-linked recessive disorder caused by mutations in the dystrophin gene affecting an estimated 1:5000 newborn males[1].
- Patients experience progressive muscle wasting, increased creatine kinase (CK) levels, difficulty controlling movement, respiratory failure and heart failure.

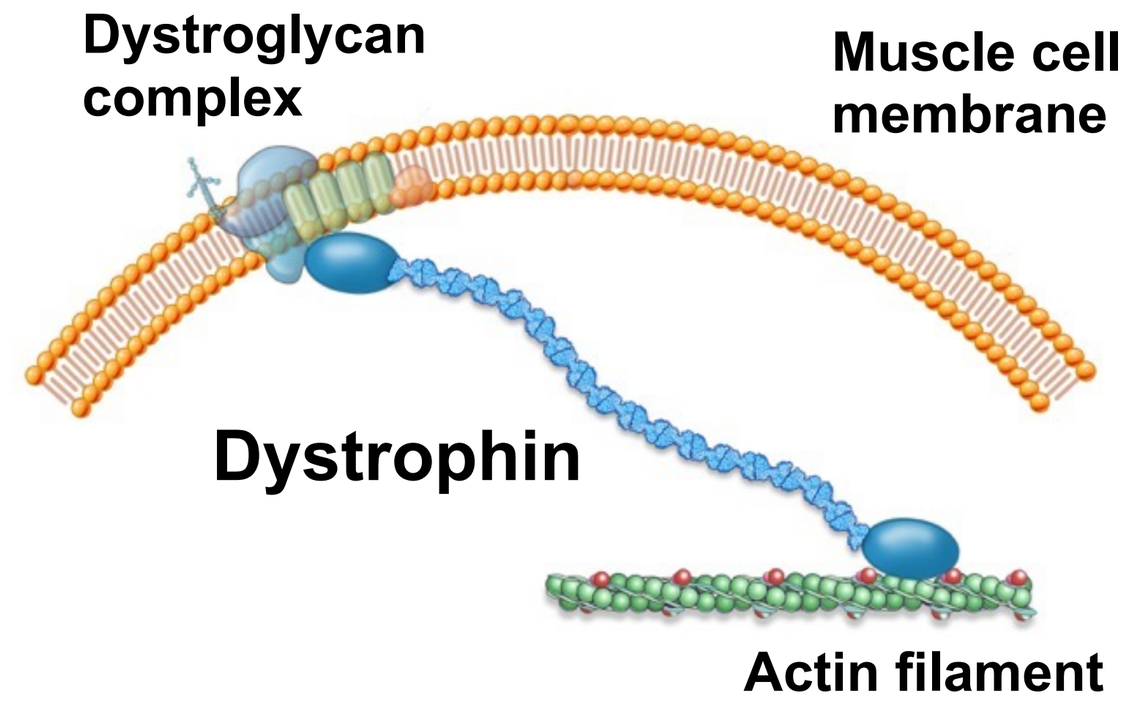
Dystroglycan complex
Muscle cell membrane
Dystrophin
Actin filament

- Most DMD-causing mutations are large mRNA-frameshifting deletions.
- Exon skipping with ASOs can restore the reading frame and lead to the production of a **nearly full-length** dystrophin protein which **maintains functional domains**; however, clinical activity is limited by the poor durability and biodistribution of ASOs.

[1] Crisafulli et al. Orphanet Journal of Rare Disease 2020.

Exon skipping with scAAV-U7 snRNAs

Vectorized U7 snRNAs combine the delivery advantages of gene therapy with the therapeutic potential of exon skipping.

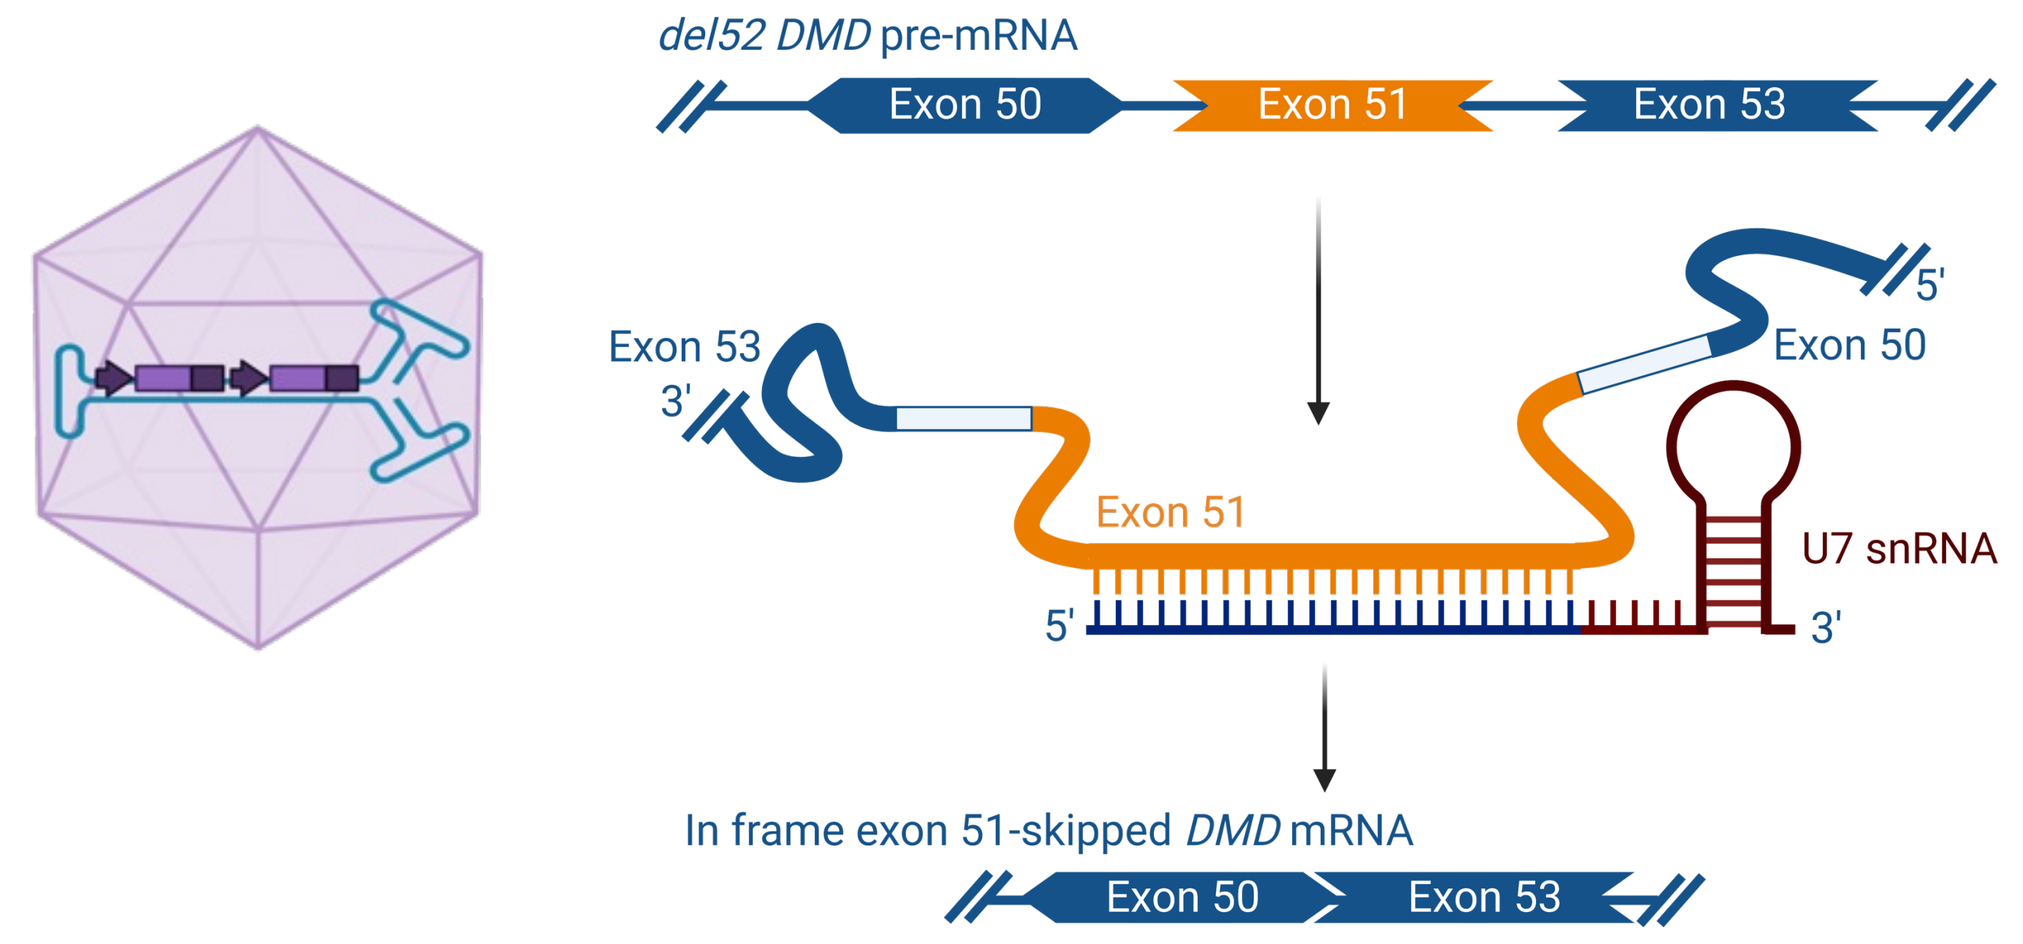
del52 DMD pre-mRNA
Exon 50
Exon 51
Exon 53
Exon 53
3'
Exon 50
5'
Exon 51
U7 snRNA
5'
3'
In frame exon 51-skipped DMD mRNA
Exon 50
Exon 53

LOCANAbio

- ✓ **Generates functional, near full-length dystrophin**
- ✓ **Maintains normal tissue expression profile of dystrophin**
- ✓ **Single administration of AAV with durable effect**
- ✓ **Non-Immunogenic**
- ✓ **Can target multiple splicing regulatory sites**
- ✓ **Ideally suited for Bespoke Gene Therapy Consortium approach**
- ✓ **Clinical validation with unprecedented levels of exon 2 skipping in Dup2 DMD patients[2,3].**

[2] Megan Waldrop, ASGCT 2022. [3] Simmons et al. Mol Ther Methods Clinical Dev 2021.

Acknowledgements

These studies are supported by Locanabio, Inc. We thank the investors and colleagues that provided support for this study.

Solving AAV snRNA packaging challenges

Locanabio's proprietary packaging and manufacturing solution has opened the field of snRNAs for therapeutic applications

Conventional snRNA packaging yields truncations and product heterogeneity

Locanabio's packaging strategy yields intended full-length genomes

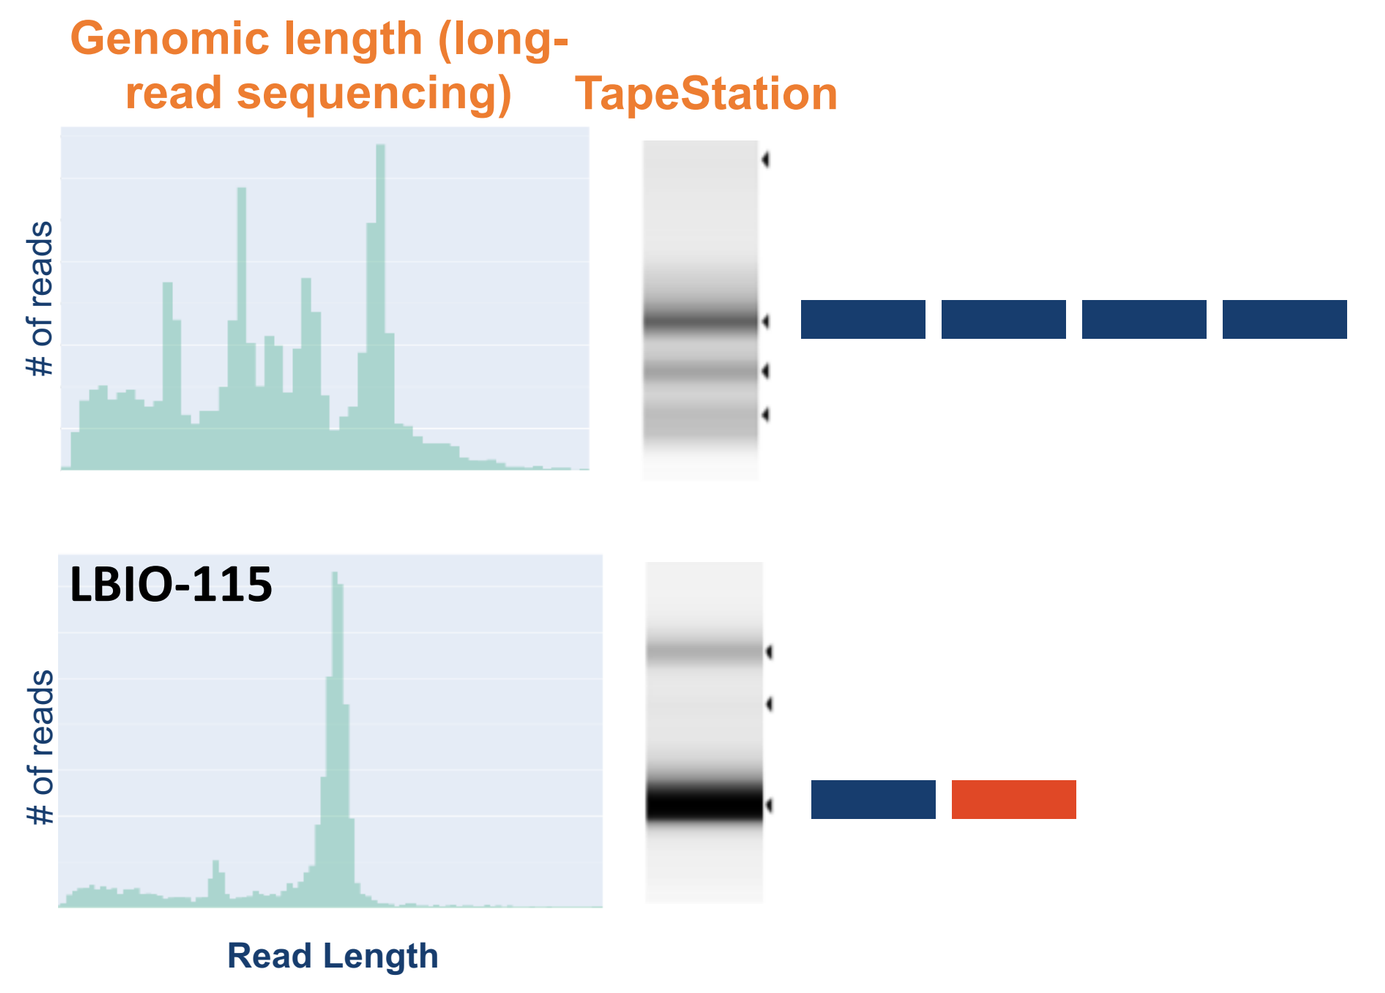
Genomic length (long-read sequencing)
TapeStation
of reads
LBIO-115
of reads
Read Length

Exon 51 skipping and dystrophin restoration in human DMD cells *in vitro*

Exon 51 skipping and dystrophin restoration after LBIO-115 treatment

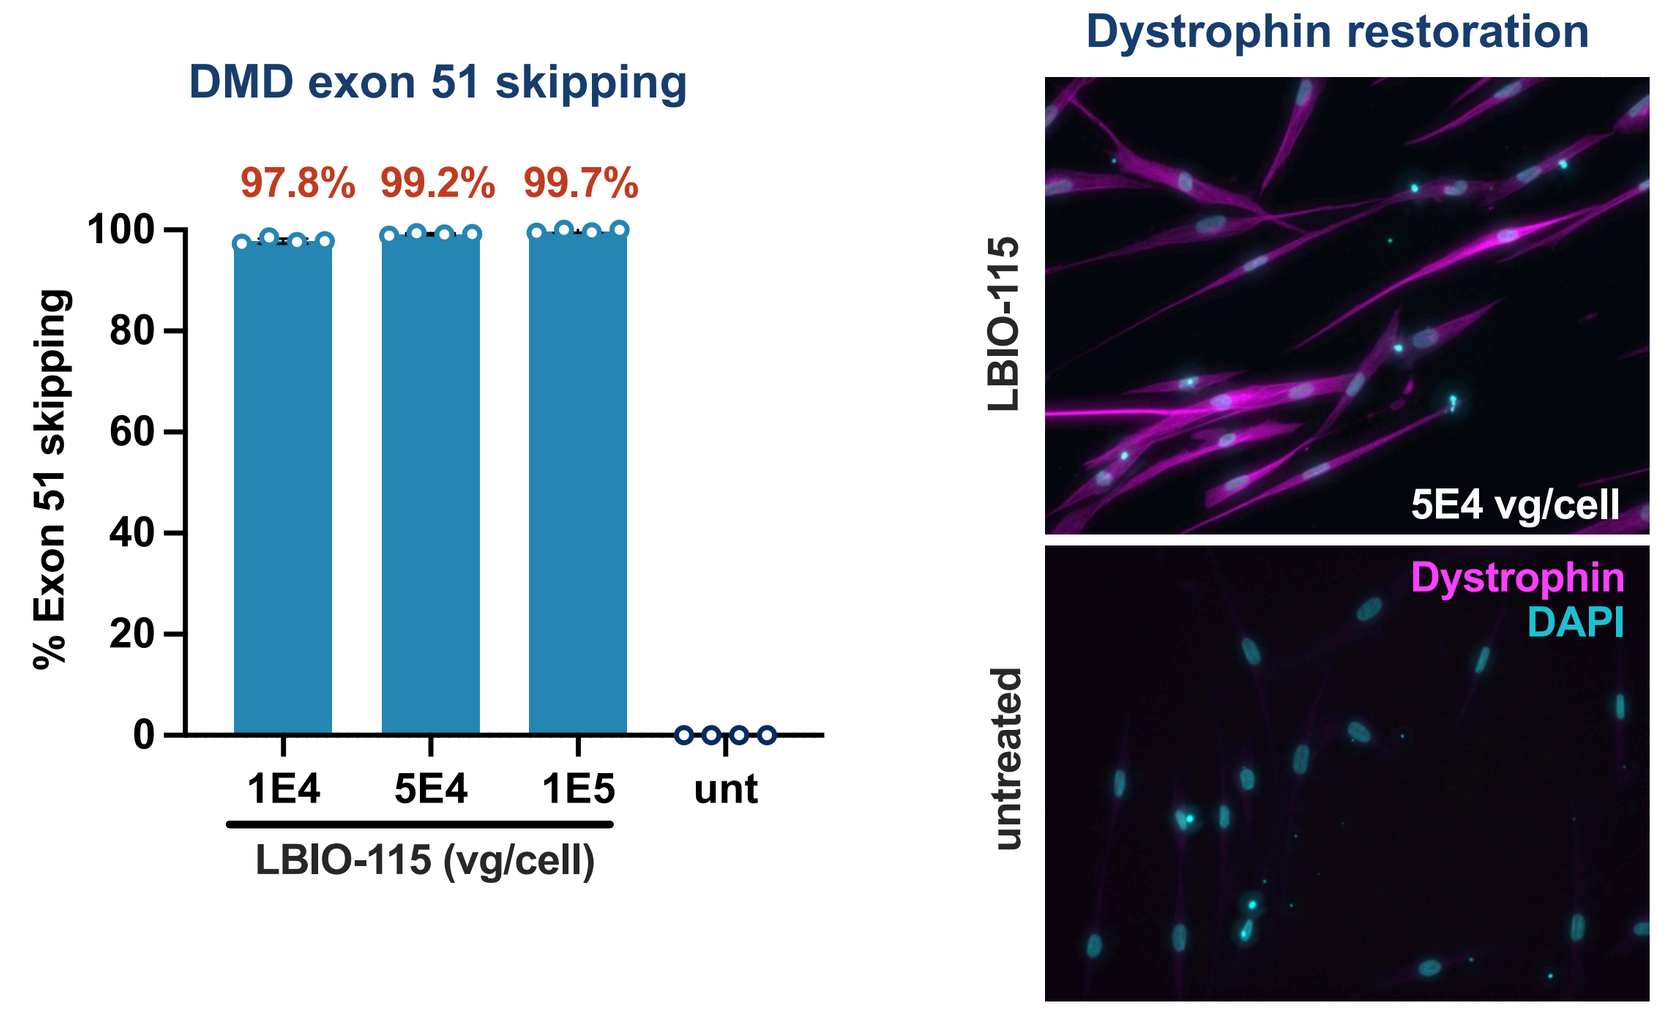
DMD exon 51 skipping
97.8%
99.2%
99.7%
% Exon 51 skipping
1E4
5E4
1E5
unt
LBIO-115 (vg/cell)
Dystrophin restoration
LBIO-115
5E4 vg/cell
untreated
Dystrophin
DAPI

Dystrophin restoration and nNos localization in human DMD cardiomyocytes after LBIO-115 treatment

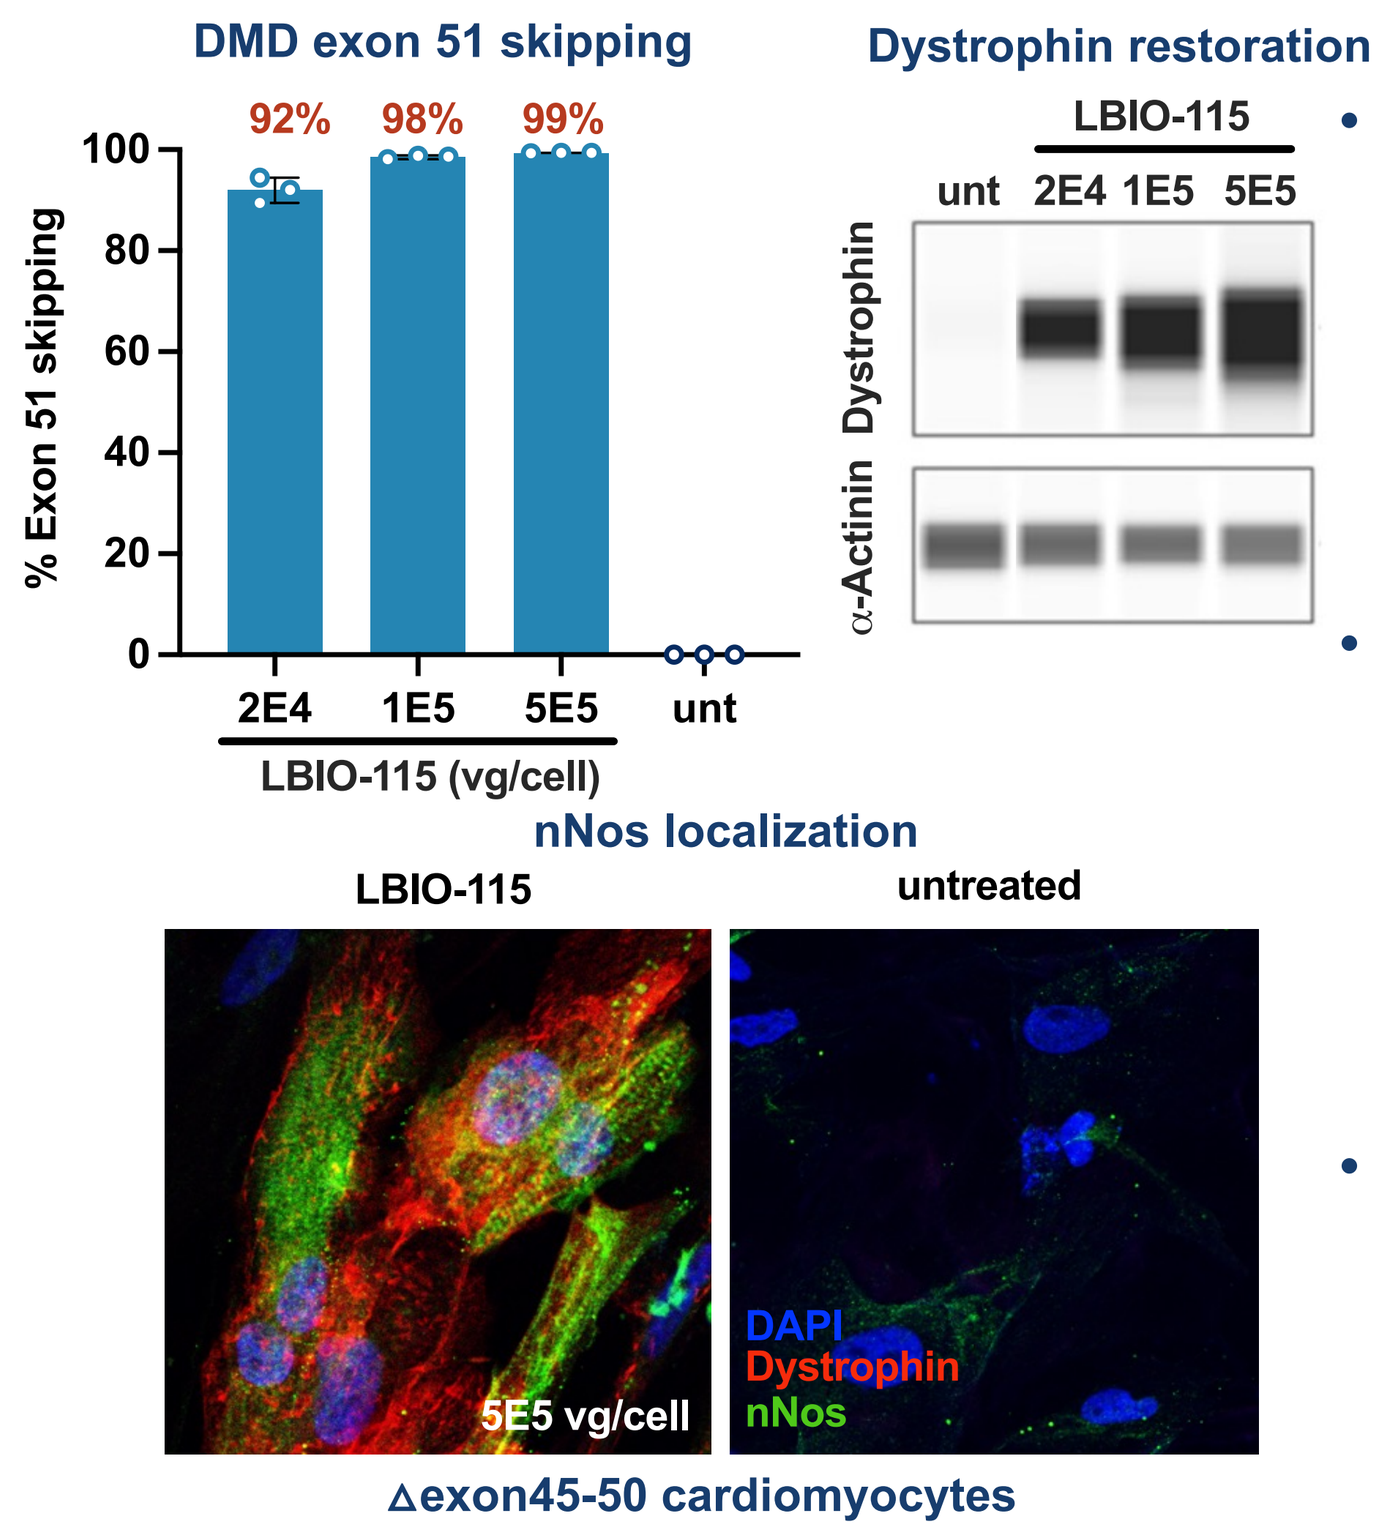
DMD exon 51 skipping
92%
98%
99%
% Exon 51 skipping
2E4
1E5
5E5
unt
LBIO-115 (vg/cell)
Dystrophin restoration
LBIO-115
unt 2E4 1E5 5E5
Dystrophin
α-Actinin
nNos localization
LBIO-115
untreated
5E5 vg/cell
DAPI
Dystrophin
nNos
Δexon45-50 cardiomyocytes

- Neuronal nitric oxide synthase (nNos) localizes to the subsarcolemma by binding to to dystrophin and its mis-localization is implicated in the pathology of DMD.
- nNos localization is dependent on R16-17 and CT functional domains within the dystrophin protein which may not be included in microdystrophin gene therapies.
- LBIO-115 leads to restoration of nearly full-length dystrophin as well as nNos localization in cardiomyocytes.

Exon 51 skipping, dystrophin restoration and improved muscle health *in vivo*

Robust exon 51 skipping 4 weeks post-intramuscular treatment with LBIO-115 in del52hDMD/mdx mouse model

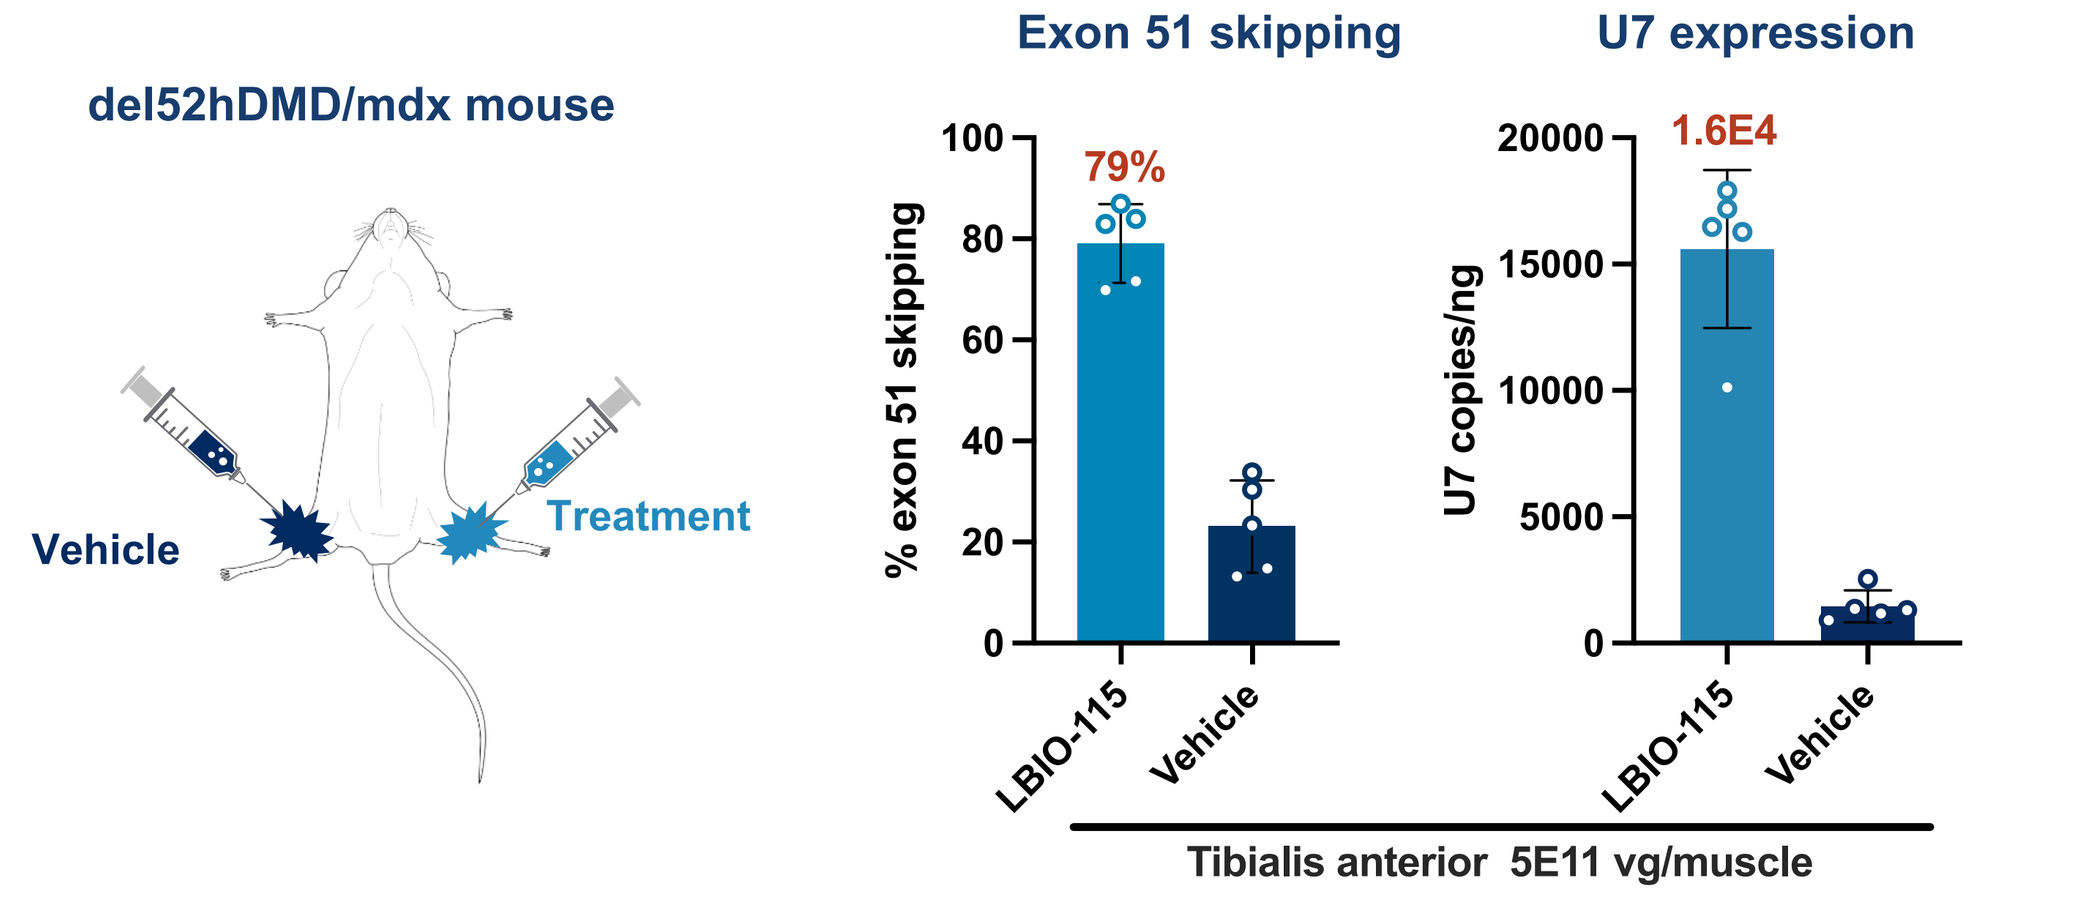
del52hDMD/mdx mouse
Vehicle
Treatment
Exon 51 skipping
79%
% exon 51 skipping
LBIO-115
Vehicle
U7 expression
1.6E4
U7 copies/ng
Tibialis anterior 5E11 vg/muscle

Dystrophin protein restoration 4 weeks post-IM treatment

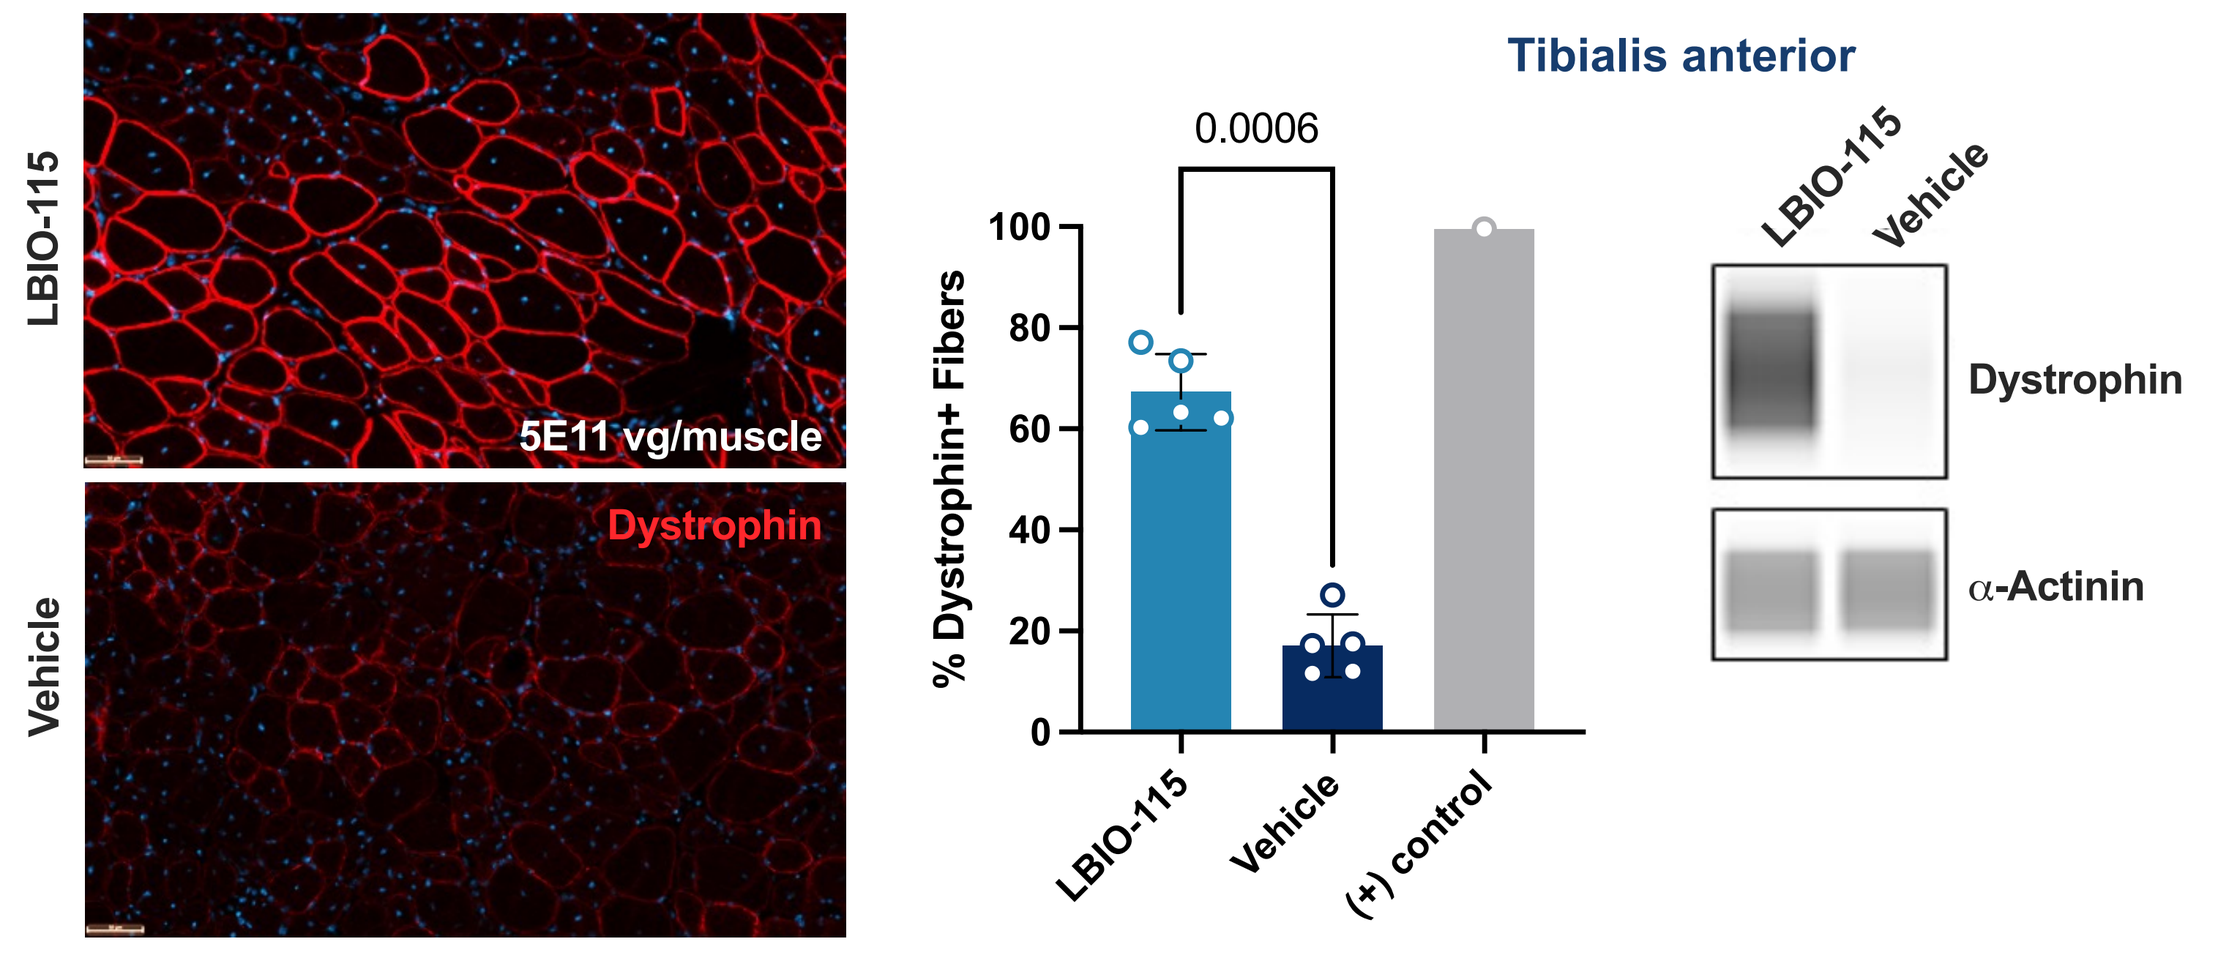
LBIO-115
5E11 vg/muscle
Vehicle
Dystrophin
Tibialis anterior
0.0006
% Dystrophin+ Fibers
LBIO-115
Vehicle
(+) control
Dystrophin
α-Actinin

Improved DMD muscle pathology 4 weeks post-IM treatment

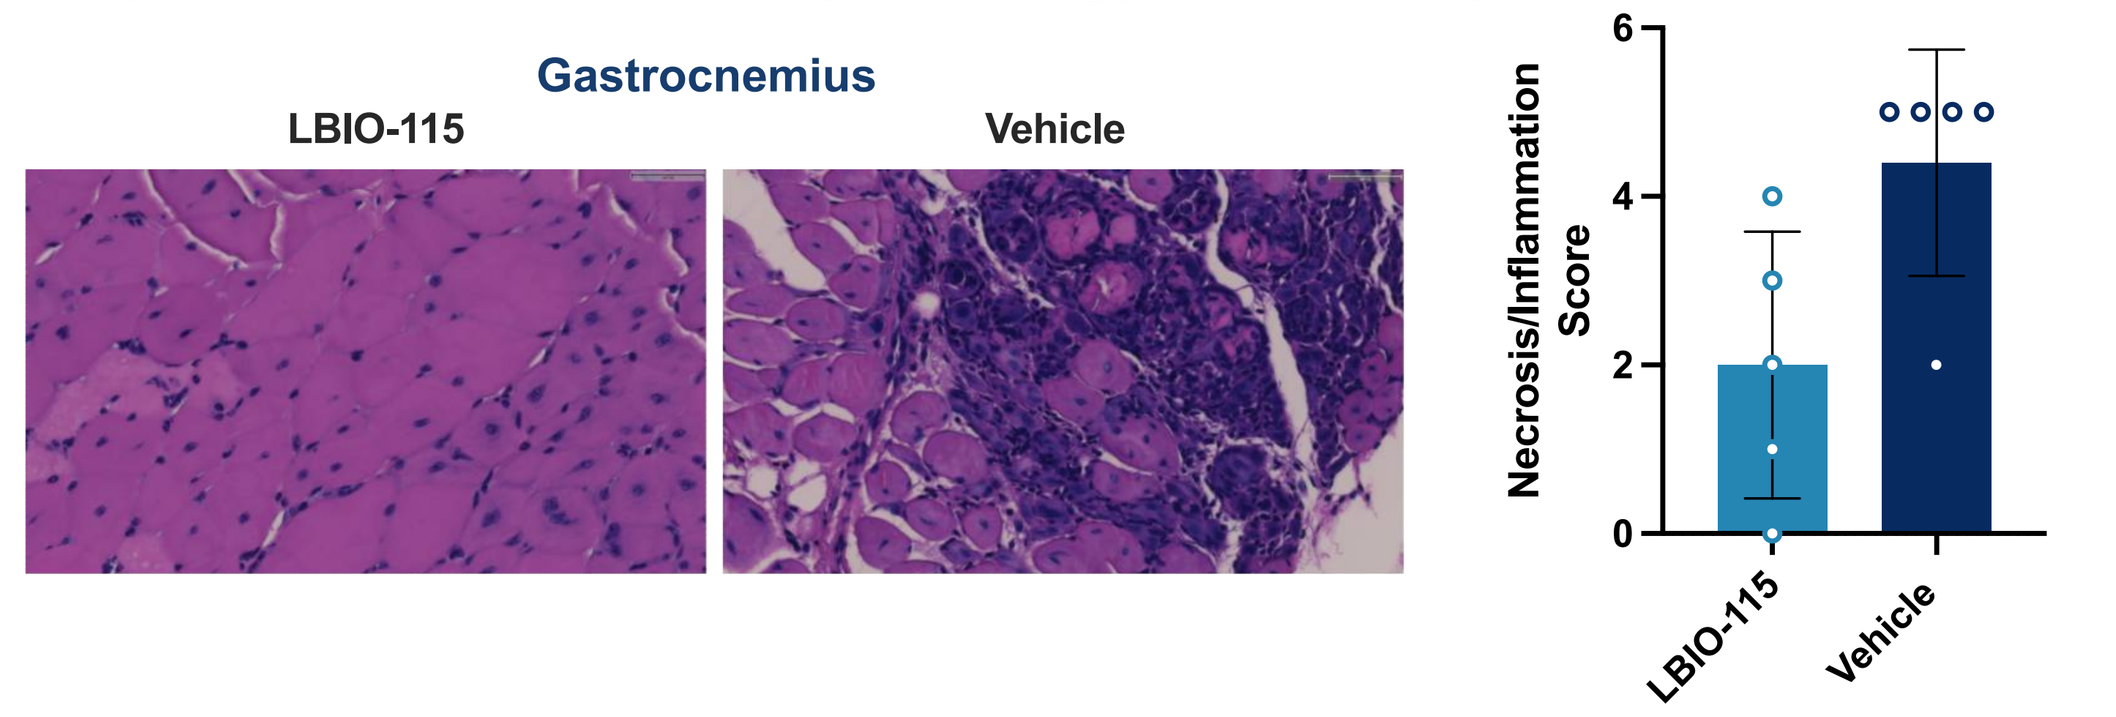
Gastrocnemius
LBIO-115
Vehicle
Necrosis/Inflammation Score

IV delivery of LBIO-115 results in robust exon skipping across tissues

Exon 51 skipping correlates with transgene expression 3-4 weeks following LBIO-115 IV administration

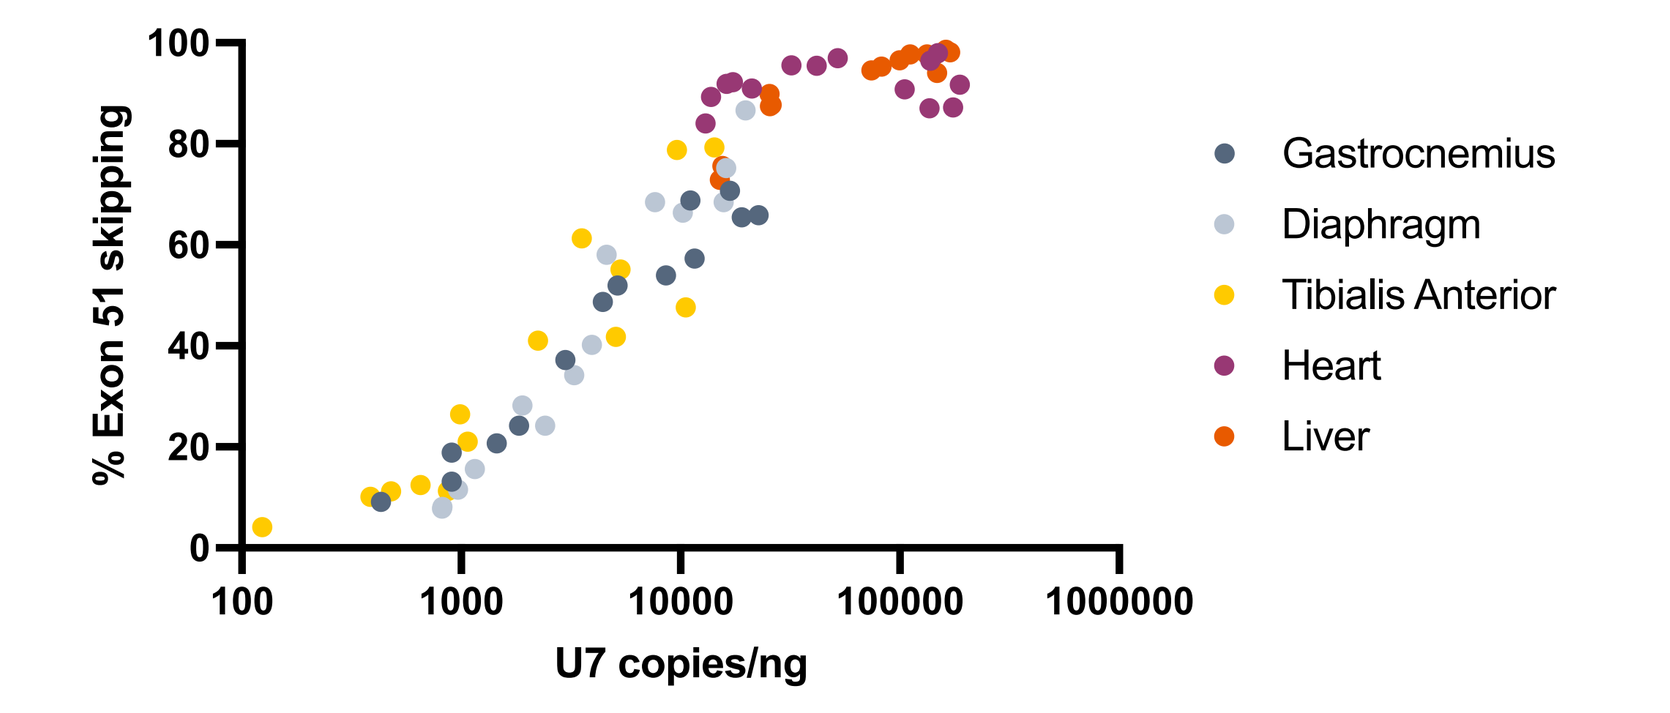
% Exon 51 skipping
U7 copies/ng
Gastrocnemius
Diaphragm
Tibialis Anterior
Heart
Liver

Dose-dependent, robust dystrophin protein restoration 3-4 weeks post-IV treatment with LBIO-115

Tissue	% Dystrophin restoration*		
	2.6E13 vg/kg	8.8E13 vg/kg	2.5E14 vg/kg**
Heart	57.0%	92.7%	79.6%
Diaphragm	7.22%	14.6%	28.3%
Gastrocnemius	3.1%	14.0%	21.4%

* Normalized to dystrophin from respective C57BL/6 mouse tissues with MANEX59B antibody
** 3-week in-life

Drop in DMD markers in serum show functional correction

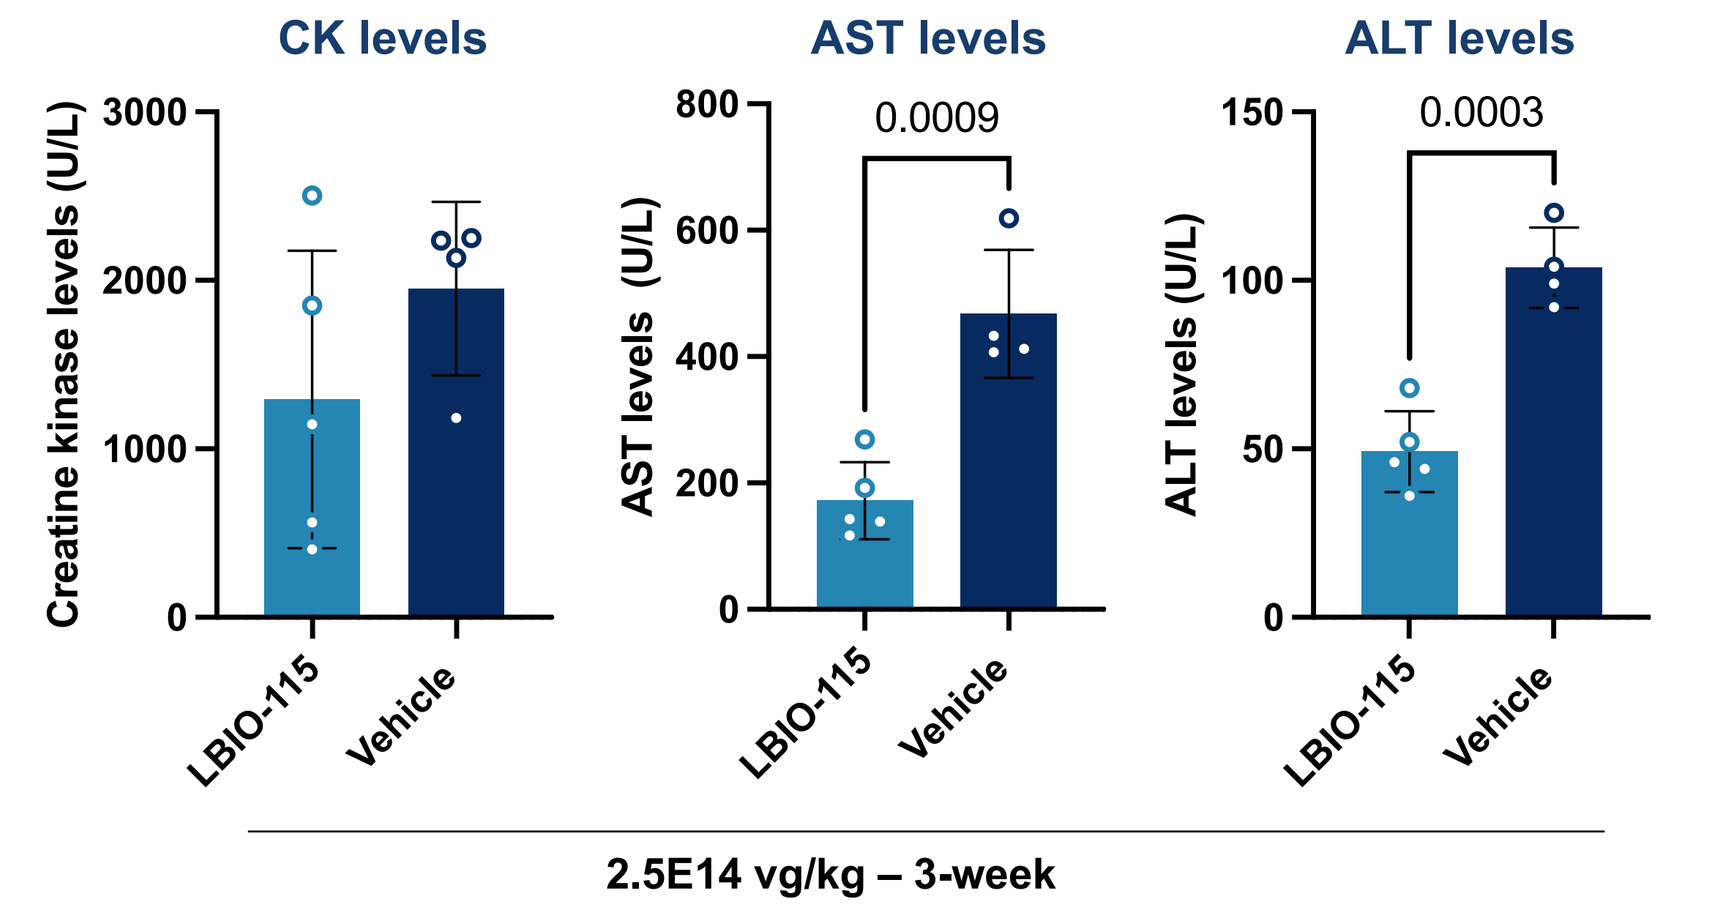
CK levels
Creatine kinase levels (U/L)
AST levels
0.0009
AST levels (U/L)
ALT levels
0.0003
ALT levels (U/L)
LBIO-115
Vehicle
2.5E14 vg/kg – 3-week

Clean off-target profile following 4-week IV LBIO-115 treatment

Off-target analysis by RNA-seq shows low differential expression in AAV9-tropic tissues

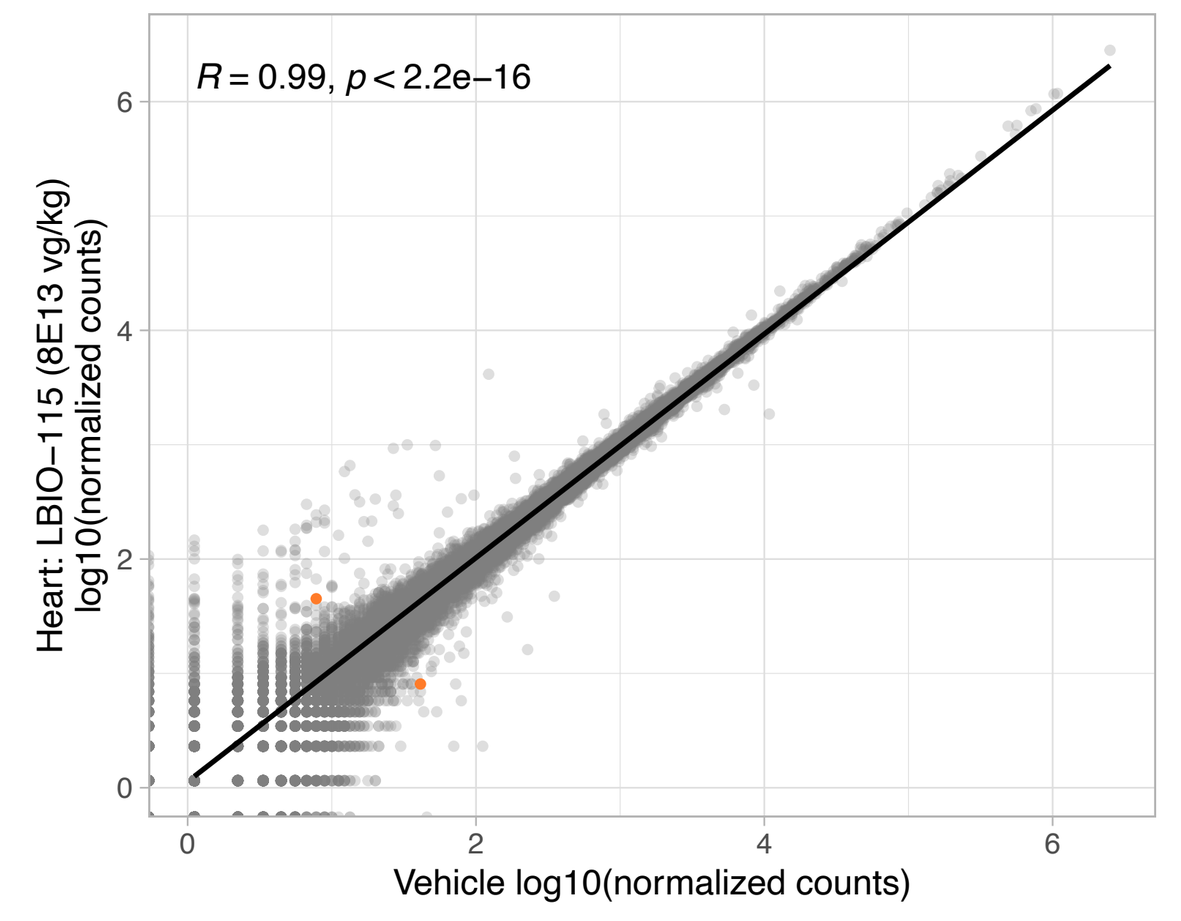
R = 0.99, p < 2.2e−16
Heart: LBIO−115 (8E13 vg/kg) log10(normalized counts)
Vehicle log10(normalized counts)

2 DETs in heart

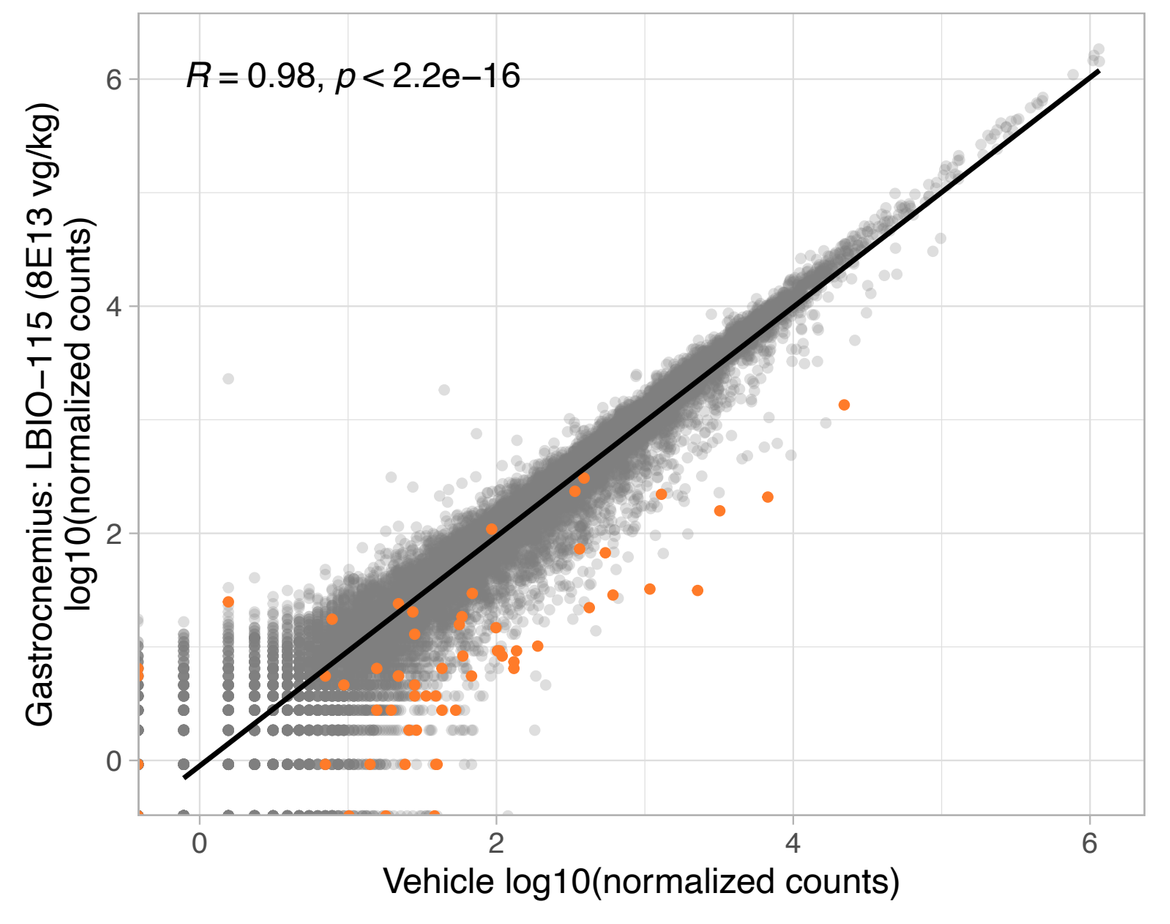
R = 0.98, p < 2.2e−16
Gastrocnemius: LBIO−115 (8E13 vg/kg) log10(normalized counts)
Vehicle log10(normalized counts)

59 DETs in Gastrocnemius

- Downregulation of inflammatory genes in skeletal muscle by GO analysis, implying improved muscle health.
- 0 DETs in liver.
- Exon 51-targeting U7 snRNA levels are ~100x lower than endogenous U1 levels 4 weeks after treatment.

Minimal alternative spliced genes

Heart: LBIO-115 (8E13 vg/kg) vs Vehicle

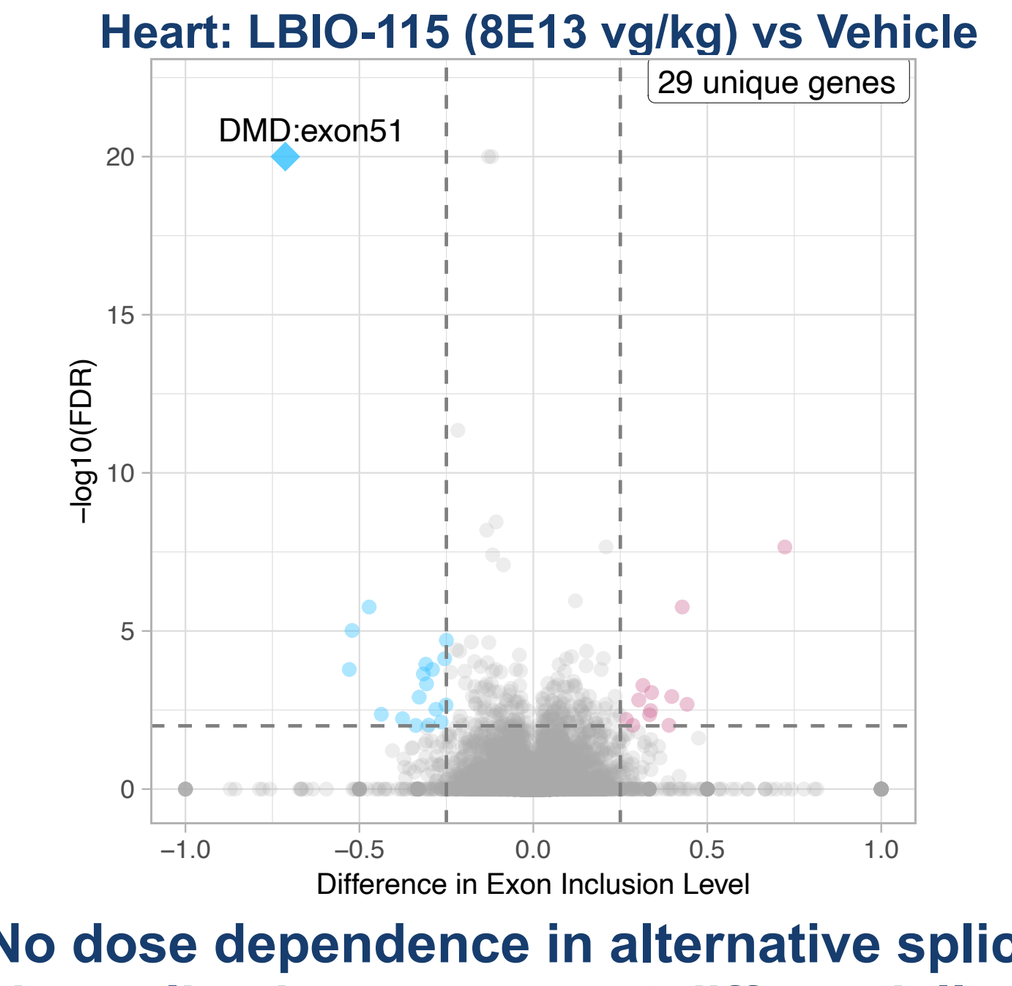
29 unique genes
DMD:exon51
−log10(FDR)
Difference in Exon Inclusion Level

No dose dependence in alternative splicing.
Alternative spliced genes are not differentially expressed.

Rapid screening of U7 antisense targeting sequences for other DMD populations

Synthetic snRNAs to screen antisense sequences targeting other therapeutically relevant exons

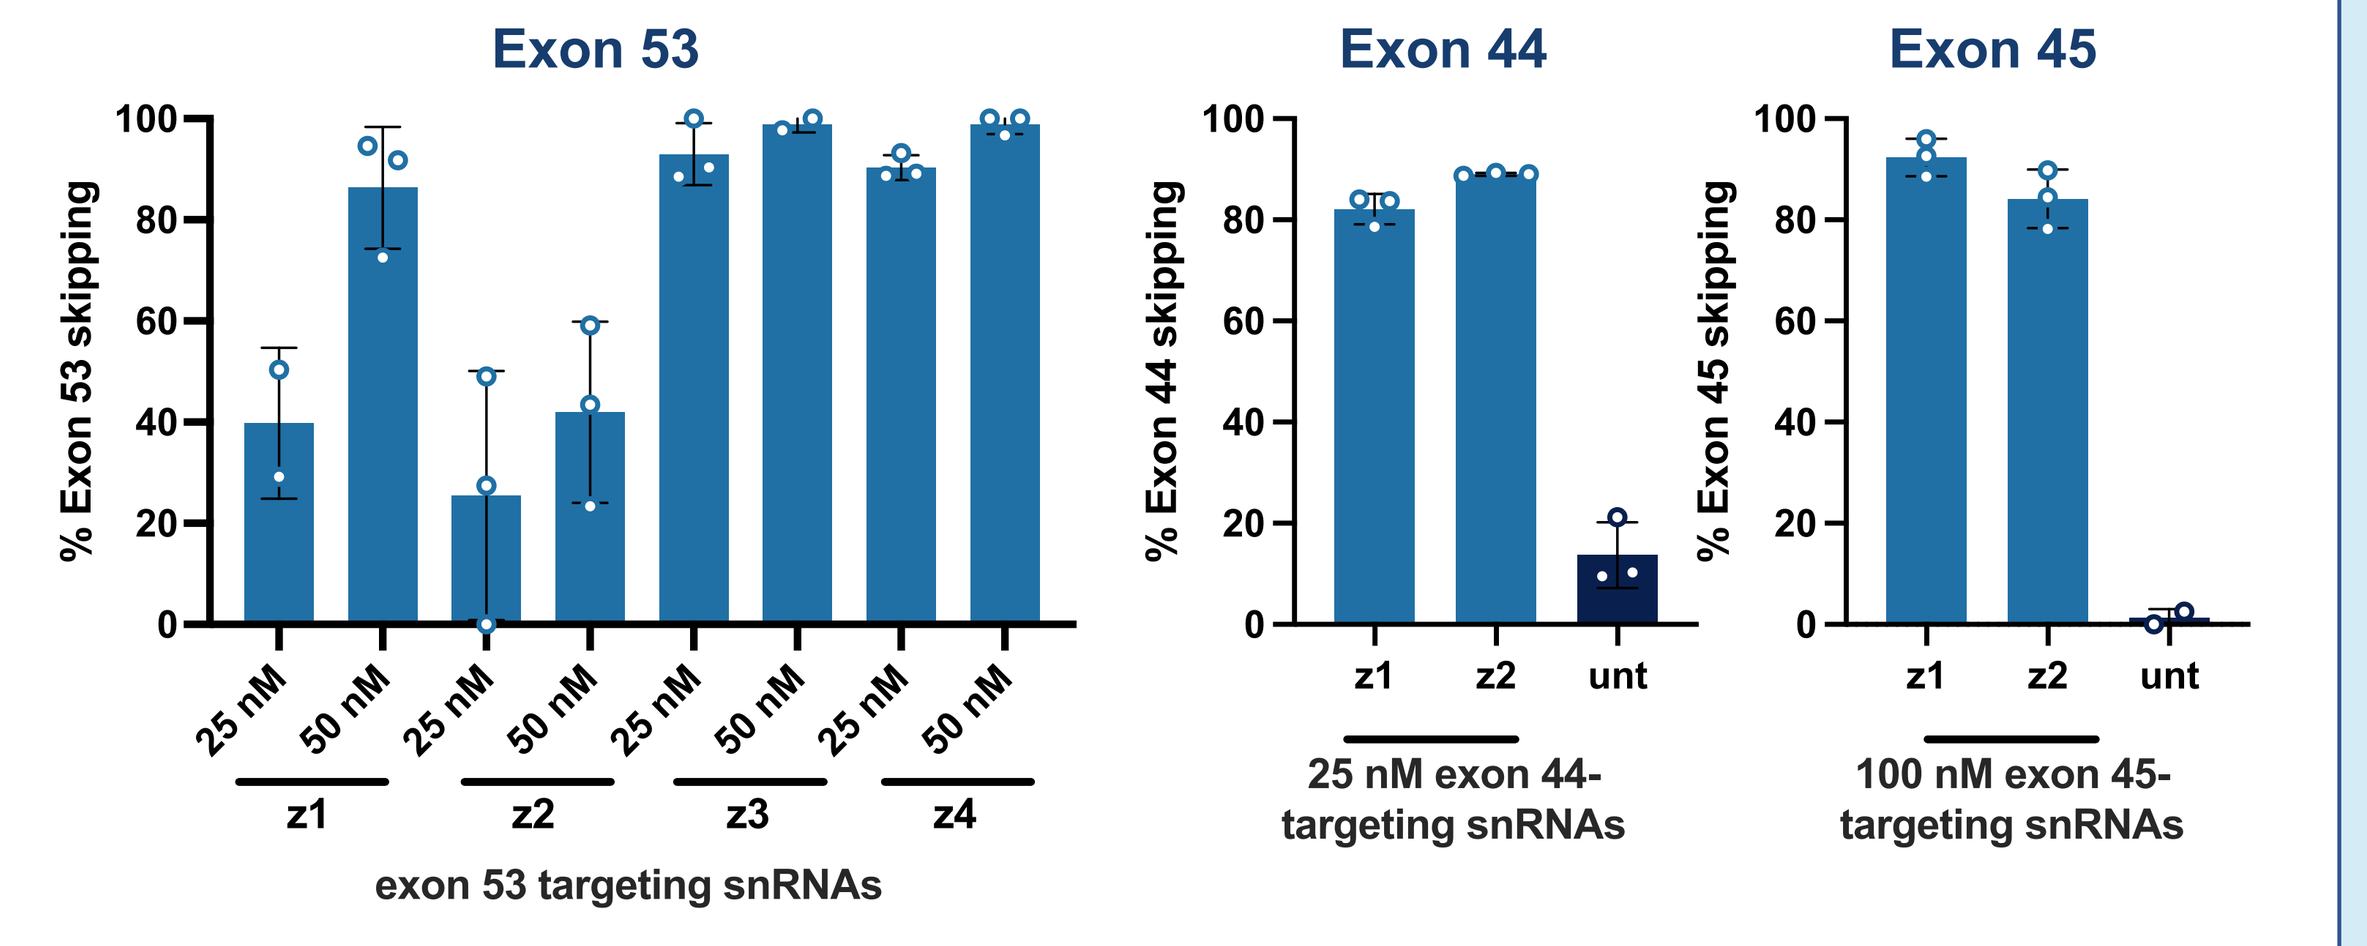
Exon 53
% Exon 53 skipping
25 nM
50 nM
z1
z2
z3
z4
exon 53 targeting snRNAs
Exon 44
% Exon 44 skipping
z1
z2
unt
25 nM exon 44-targeting snRNAs
Exon 45
% Exon 45 skipping
z1
z2
unt
100 nM exon 45-targeting snRNAs

Exon 53 skipping and dystrophin restoration with exon 53-targeting sequences packaged in Locanabio's scAAV backbone (A05014)

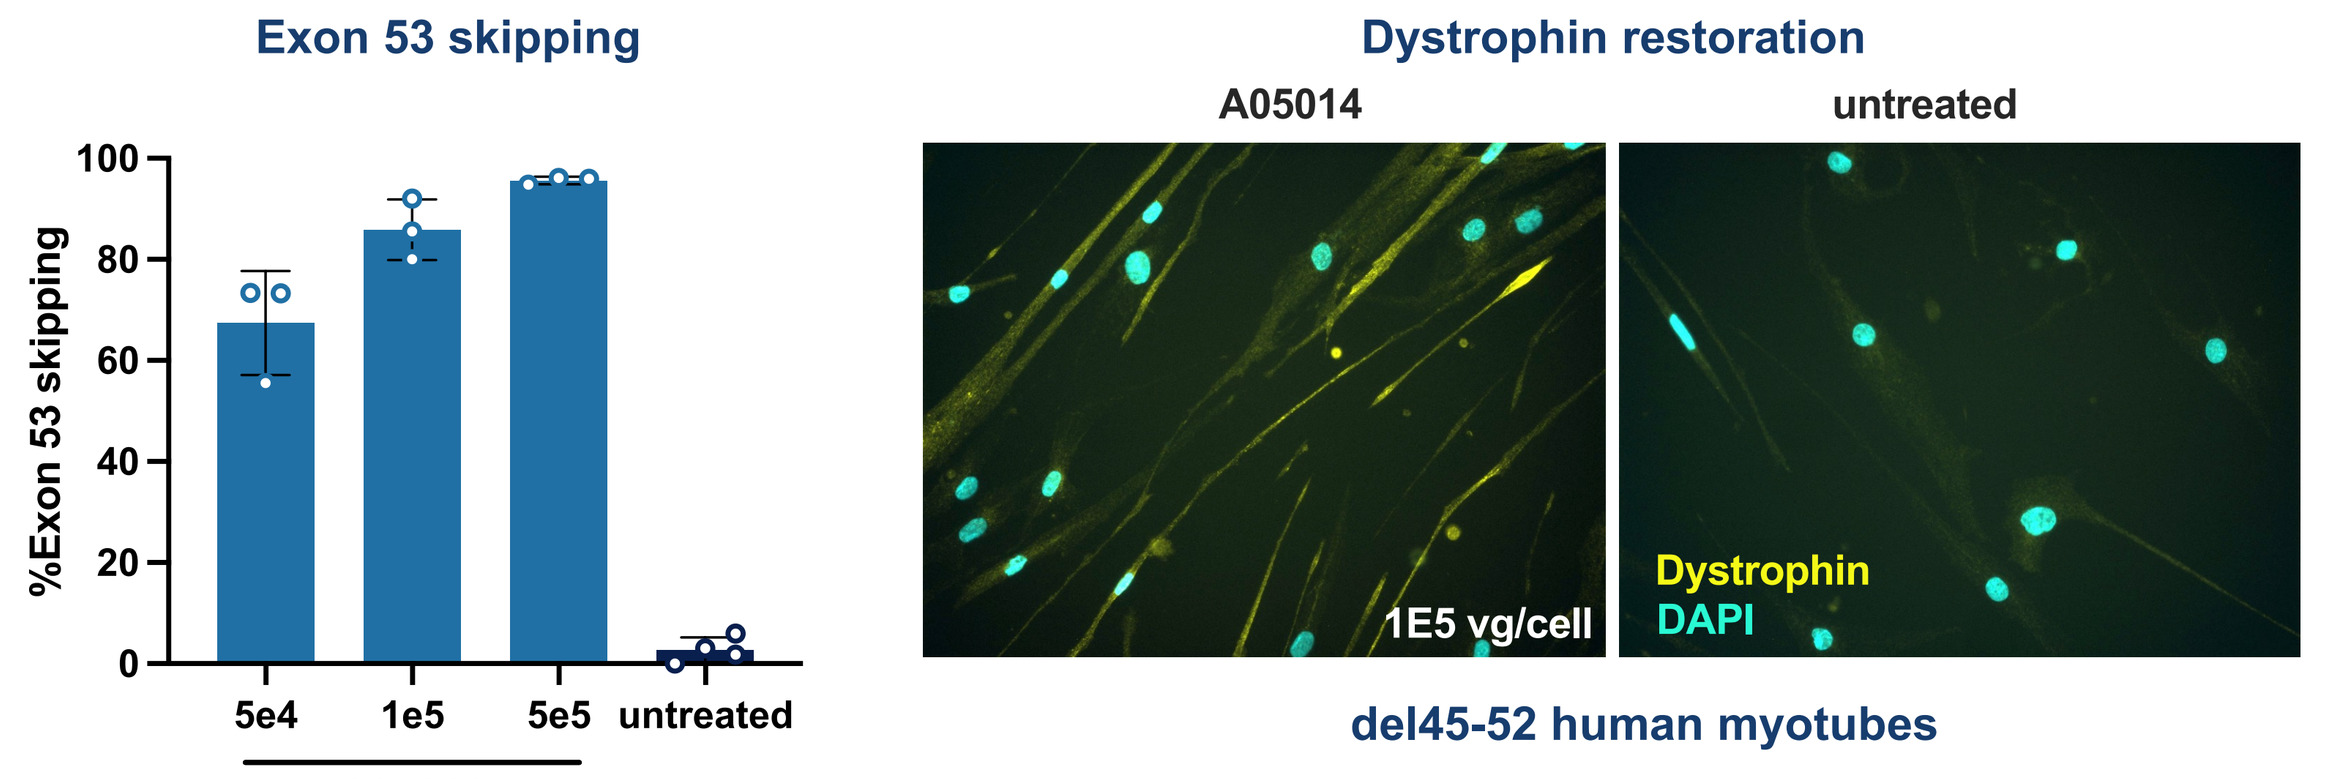
Exon 53 skipping
%Exon 53 skipping
5e4
1e5
5e5
untreated
A05014
Dystrophin restoration
A05014
untreated
1E5 vg/cell
Dystrophin
DAPI
del45-52 human myotubes

***In vivo* efficacy and expression from A05014 support rapid therapeutic development for skipping other *DMD* exons**

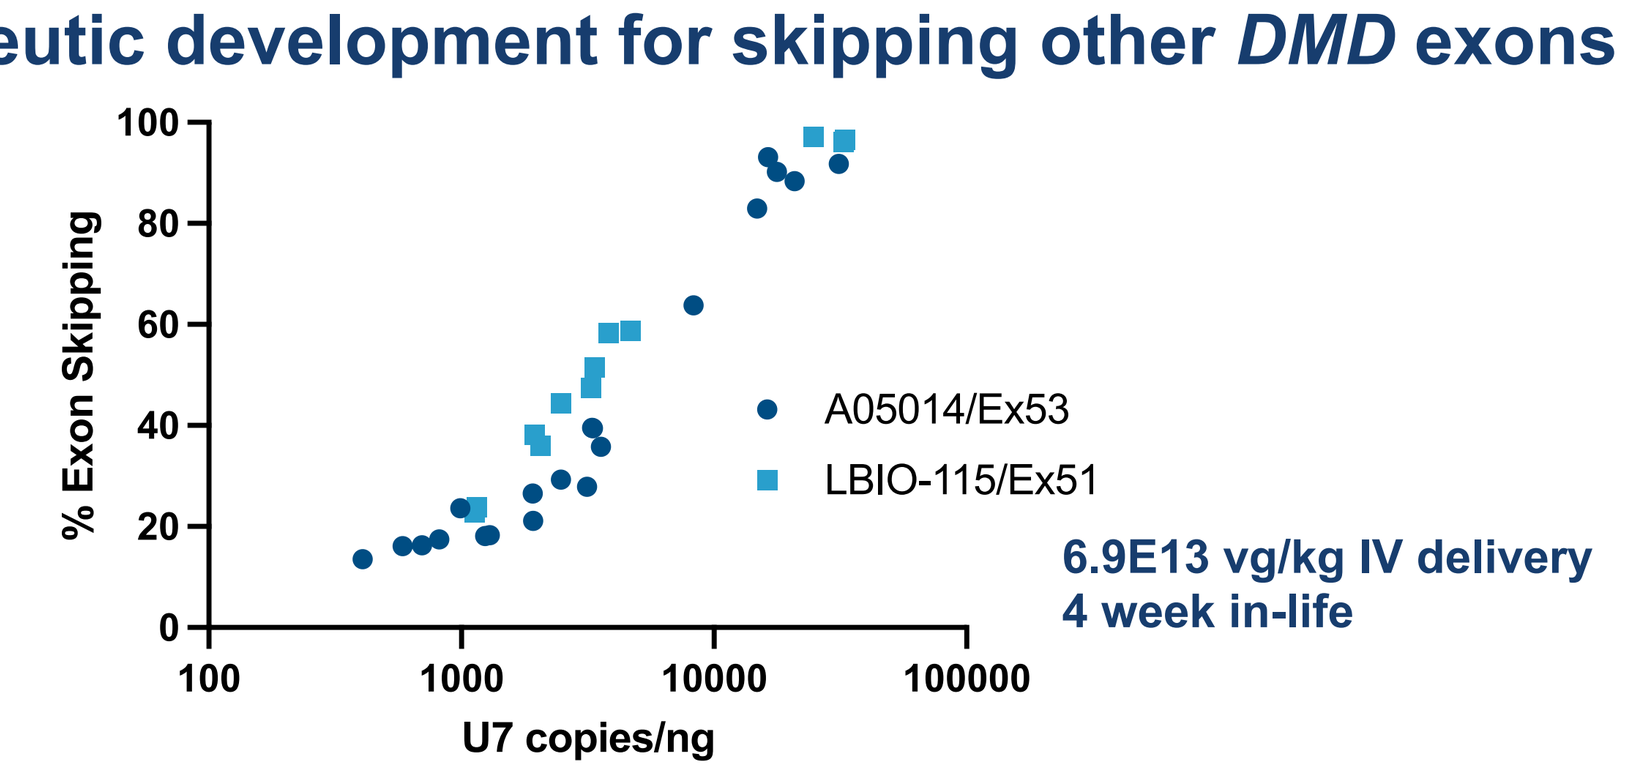
% Exon Skipping
U7 copies/ng
A05014/Ex53
LBIO-115/Ex51
6.9E13 vg/kg IV delivery
4 week in-life

Conclusions

- Full-length packaging of intended scAAV U7-expressing genome.
- Near-complete exon 51 skipping accompanied by dystrophin restoration and nNos localization in human DMD cell models.
- Exon skipping, dystrophin restoration and improved muscle health demonstrated in d52hDMD/mdx mice with LBIO-115 treatment.
- Exon 51 skipping and dystrophin restoration correlated with snRNA expression across tissues following IV delivery after 4 weeks.
 - Exon 51 skipping between 11-97% across doses and tissues
 - Dystrophin restoration between 3-92% across doses and tissues
- Clean off-target profile in AAV9-tropic tissues.
- 80% of DMD mutations are amenable to exon skipping therapies.
 - We have identified spacers for exons 51, 53, 44, 45 which would address 35% of the total DMD population.

Presented at: **ASGCT 26th Annual Meeting | May 16-20, 2023**

©2023 Locanabio, Inc. All rights reserved.